Post-Harvest Application of Bacteriophages on Beef as a Natural Intervention Against *E. coli* O157

Bert de Vegt[1], Silvia Klamert[1], Sander Witte[1], Joël van Mierlo[1]

[1]Micreos Food Safety B.V., Wageningen, The Netherlands

RECIPROCAL MEAT CONFERENCE 2018

Poster #117

phageguard

I. Introduction

Bacteriophages, or is short phages, are the natural enemy of bacteria that have evolved to specifically bind and infect their bacterial host cells (Figure 1). After infection, phages rapidly replicate, leading to lysis of the bacterial cell. This mechanism enables phages to selectively kill members of a specific bacterial genus, while leaving other bacterial communities intact. Together with the fact that phages are harmless to human, it makes them a safe and effective antibacterial agent against food pathogens.

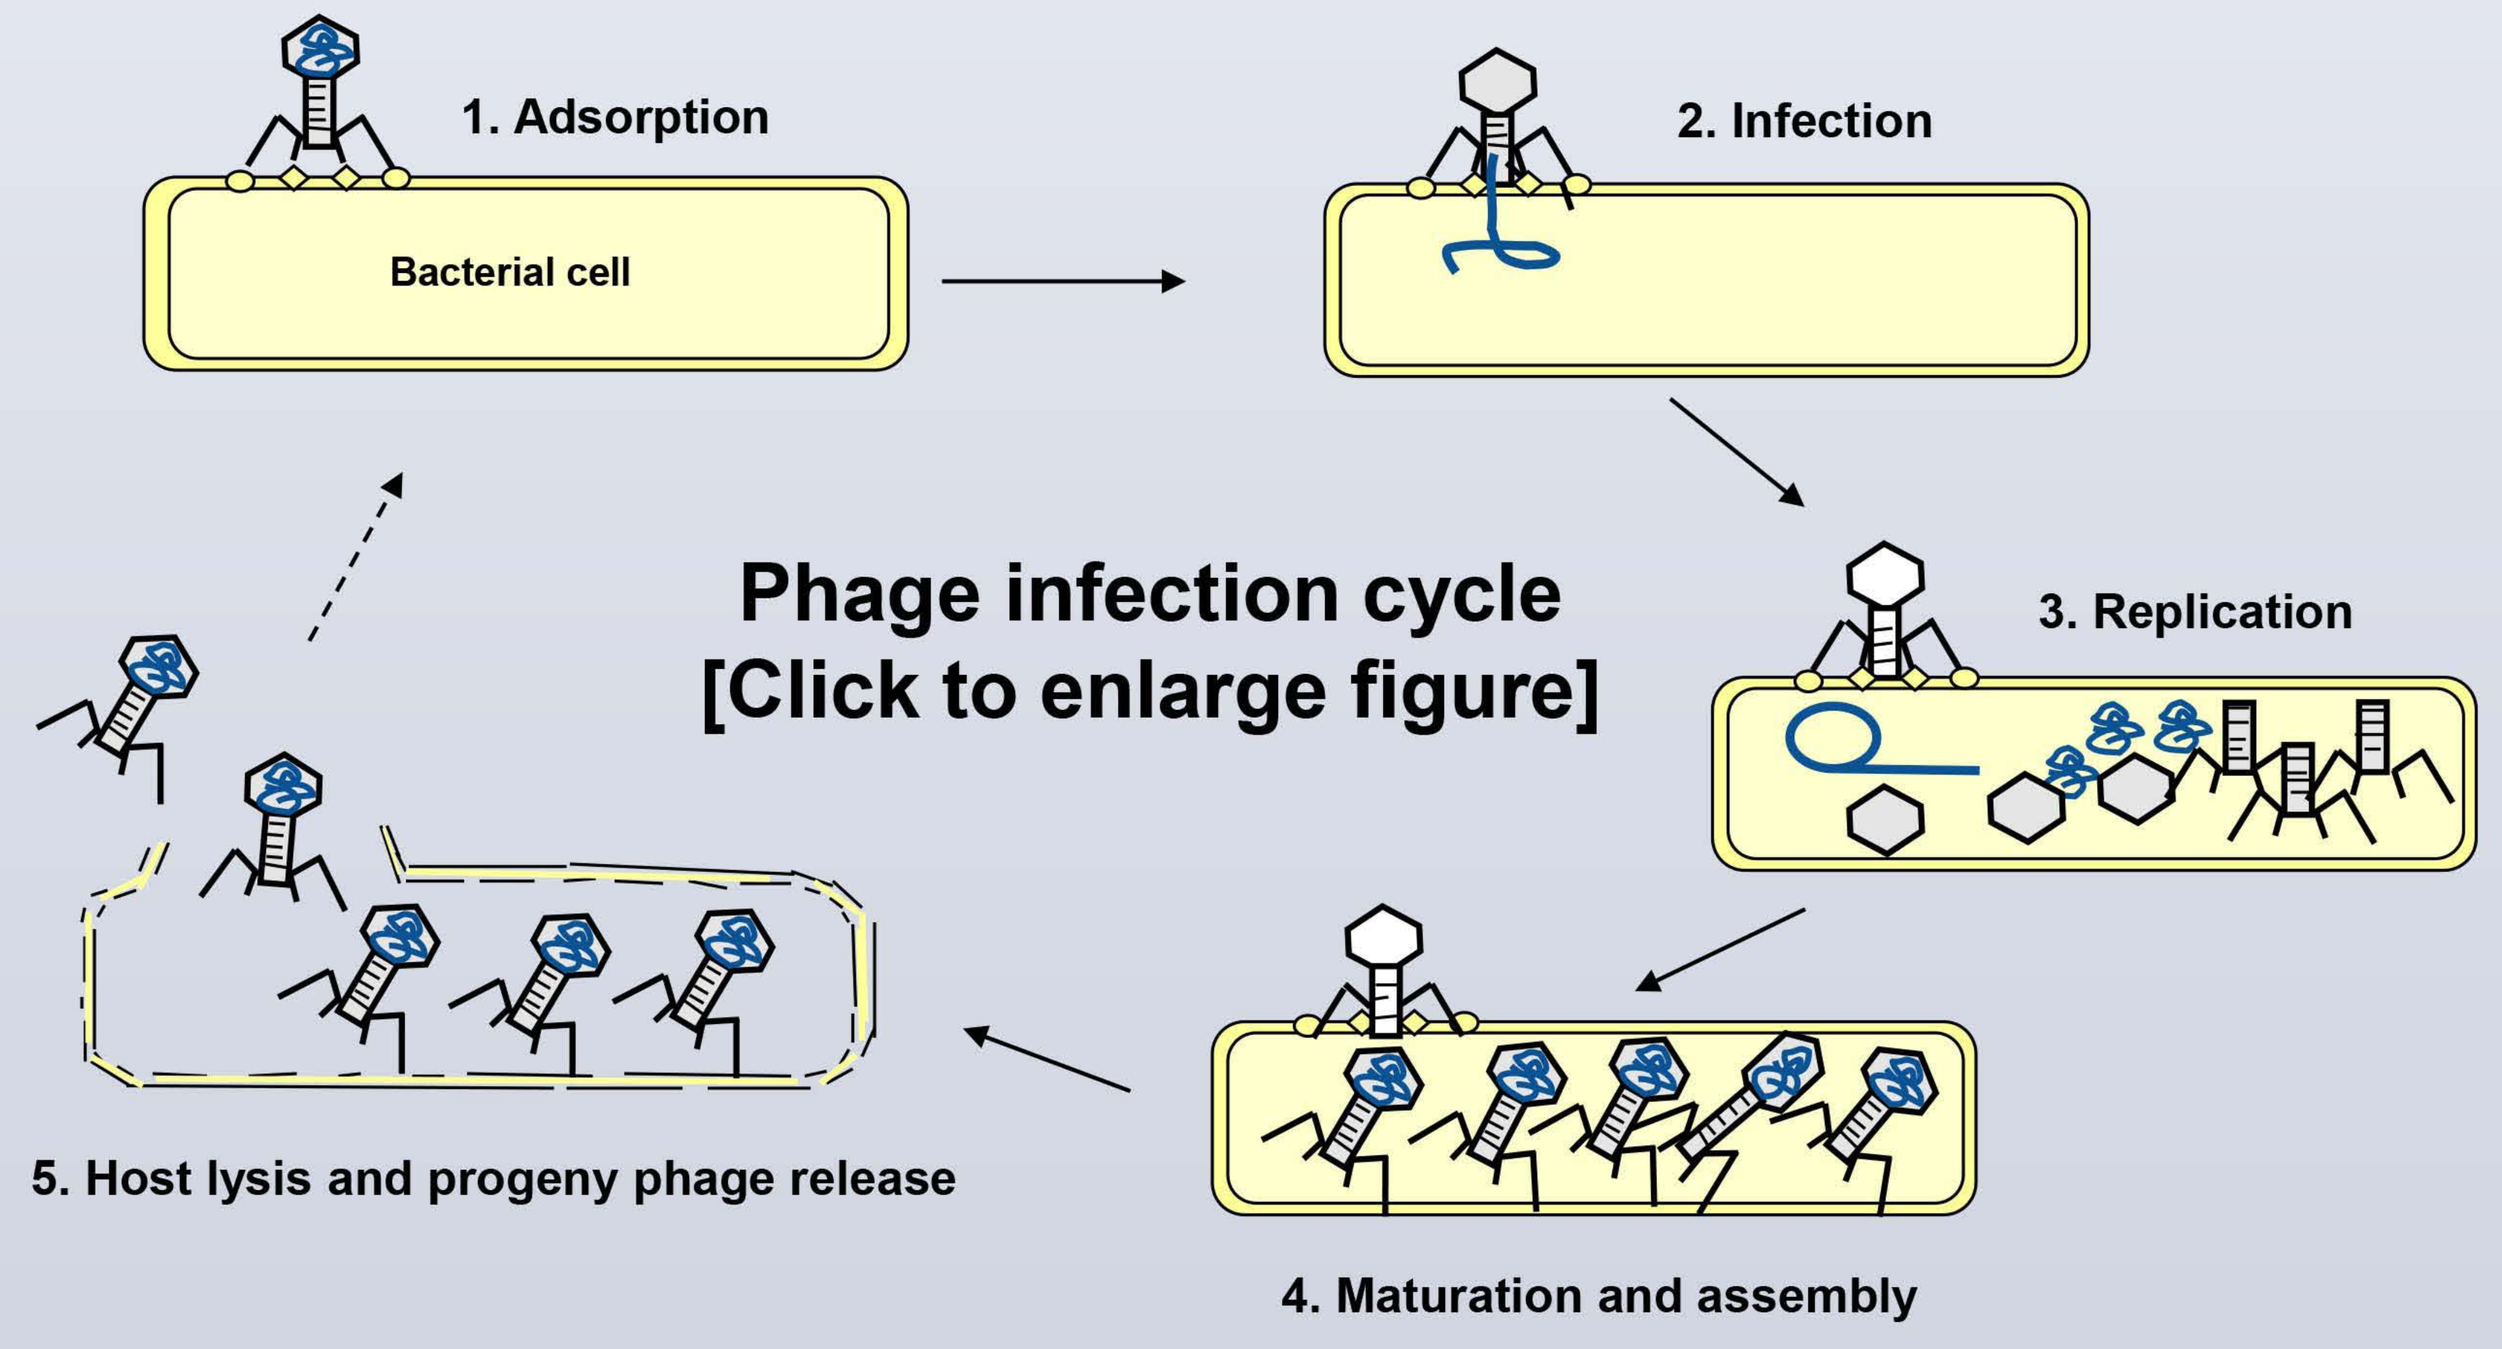
1. Adsorption
Bacterial cell
2. Infection
3. Replication
Phage infection cycle
[Click to enlarge figure]
4. Maturation and assembly
5. Host lysis and progeny phage release

Figure 1. Infection cycle of a bacteriophage. (1) After adsorption to specific receptors on the baterial host the phage (2) injects its genomic DNA into the host cell. (3) The genomic DNA starts replicating and phage components are synthesized. (4) Phages are assembled after which (5) the bacterial host lysis

III. Methods

1. Bacteriophage isolation and hostrange

Bacteriophages were isolated by adding sewage water with *E. coli* O157 culture to molten top agar (0.4%), followed by plating on an LB agar plate (1.5%). Plaques were isolated from the plates, after which phages were eluted from the plaques to obtain pure phage solutions. The isolated phages were examined for their ability to lyse 88 different *E. coli* O157 strains using the spot plate method.

2. Challenge study

Refrigerated beef samples of 3 x 3 cm were inoculated with 1×10^5 CFU/cm^2. Subsequently, samples were treated with 3×10^7 or 3×10^8 PFU/cm2 by applying 5 µL/cm2 of a cocktail containing two selected phages. Controls were treated with similar volumes of tap water. Beef samples were then incubated for 24 hours at 4 °C, after which bacteria were retrieved for enumeration at the indicated timepoints.

IV. Results (click images to enlarge)

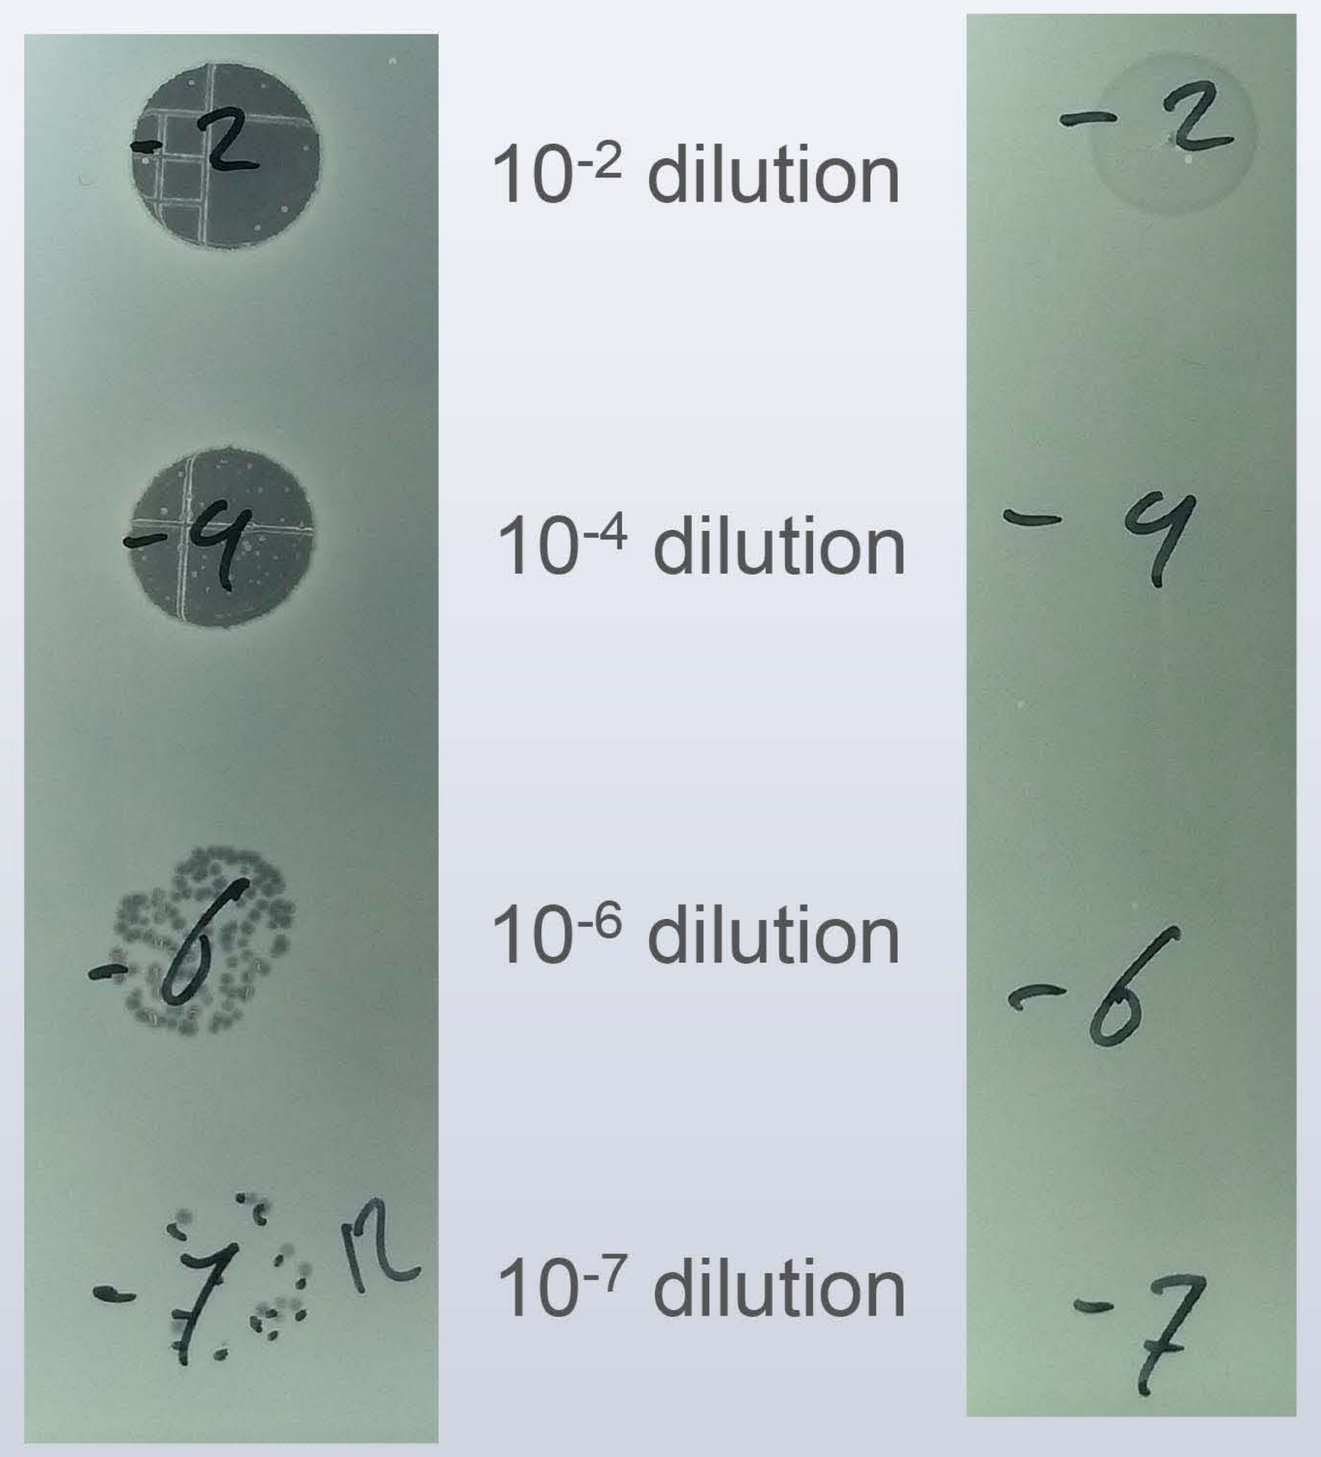
Strain = Covered
Strain = Not covered
10-2 dilution
10-4 dilution
10-6 dilution
10-7 dilution

Two phage cocktail covers 90% of all *E. coli* O157 strains

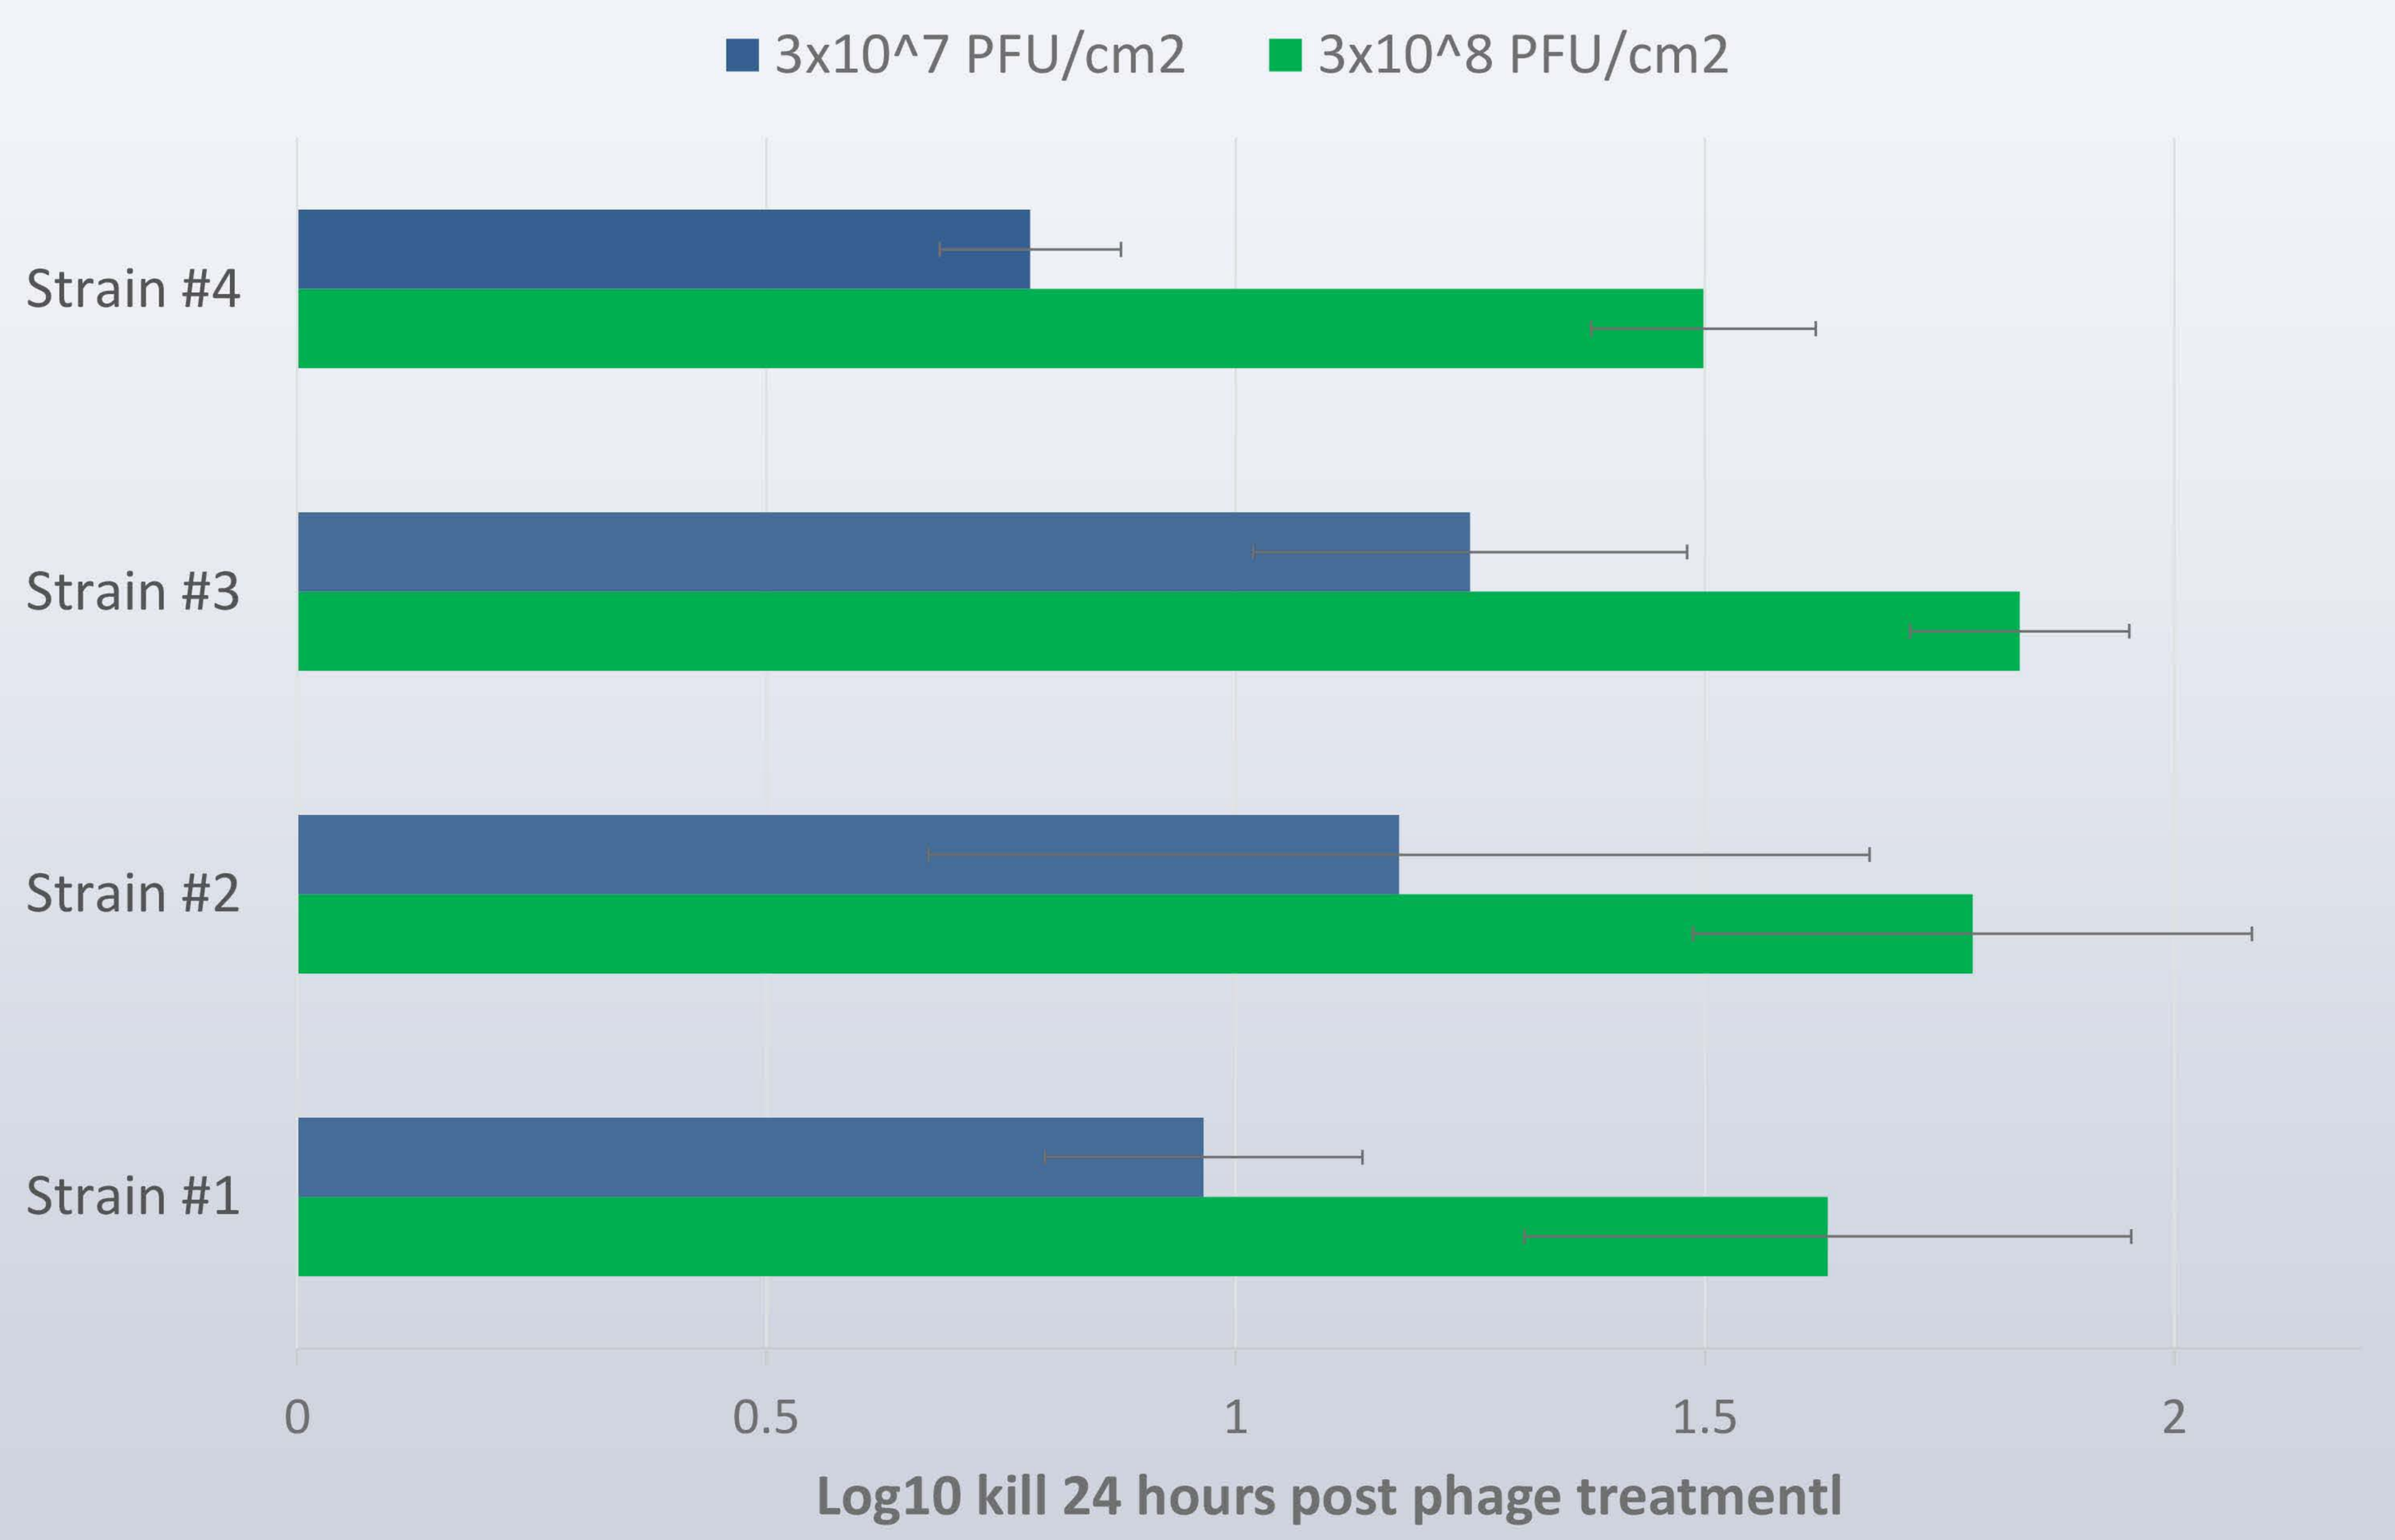
3x10^7 PFU/cm2
3x10^8 PFU/cm2
Strain #4
Strain #3
Strain #2
Strain #1
0
0.5
1
1.5
2
Log10 kill 24 hours post phage treatmentl

Phages reduce *E. coli* O157 by 0.8 – 1.9 log10

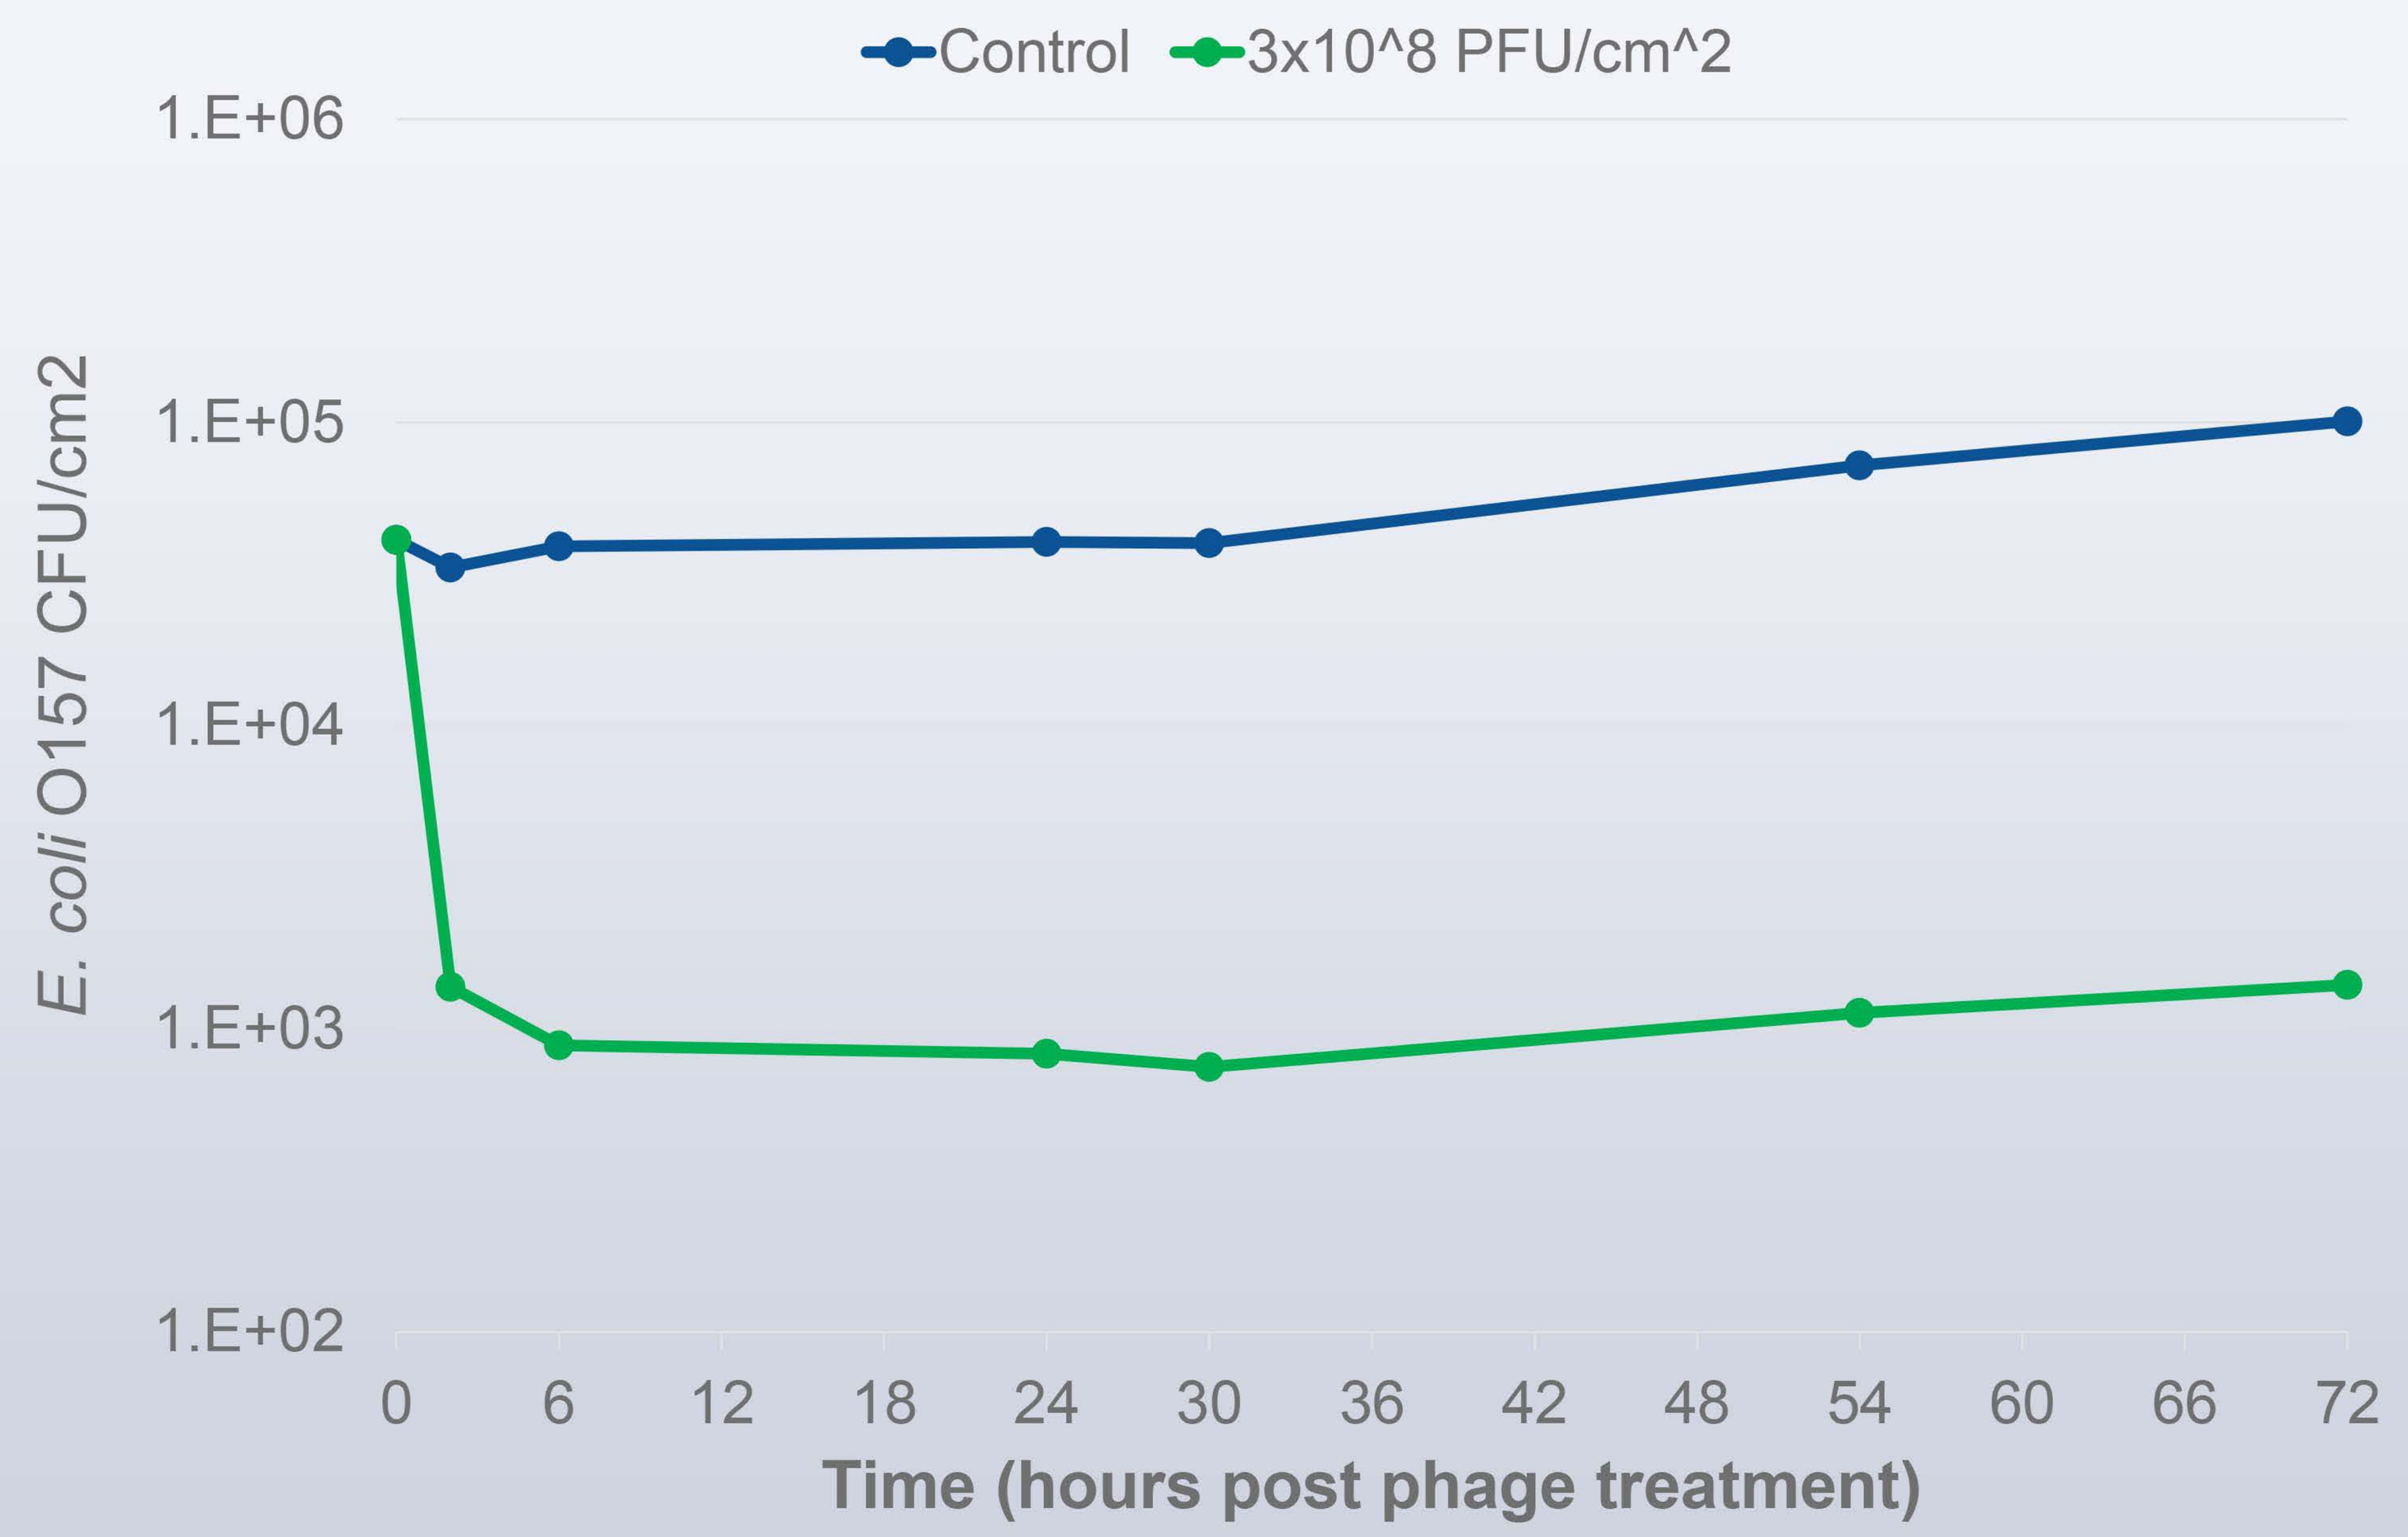
Control
3x10^8 PFU/cm^2
1.E+06
1.E+05
1.E+04
1.E+03
1.E+02
E. coli O157 CFU/cm2
0
6
12
18
24
30
36
42
48
54
60
66
72
Time (hours post phage treatment)

Maximum *E. coli* O157 kill after 6 hours

II. Objective

The objective of this study was to determine the efficacy of bacteriophages as a natural intervention against *E. coli* O157 on refrigerated beef.

V. Conclusion/Discussion

1. A two phage cocktail can cover 90% of all *E. coli* O157 strains

2. Two phage cocktail can reduce *E. coli* O157 on cold beef by up to 2 log

VI. Perspectives

GRAS status for use on beef of this two phage cocktail is expected July 2018, after which the phage cocktail is commercially available.

Post-Harvest Application of Bacteriophages on Beef as a Natural Intervention Against E. coli O157

Bert de Vegt[1], Silvia Klamert[1], Sander Witte[1], Joël van Mierlo[1]

[1]Micreos Food Safety B.V., Wageningen, The Netherlands

Poster #117

phageguard

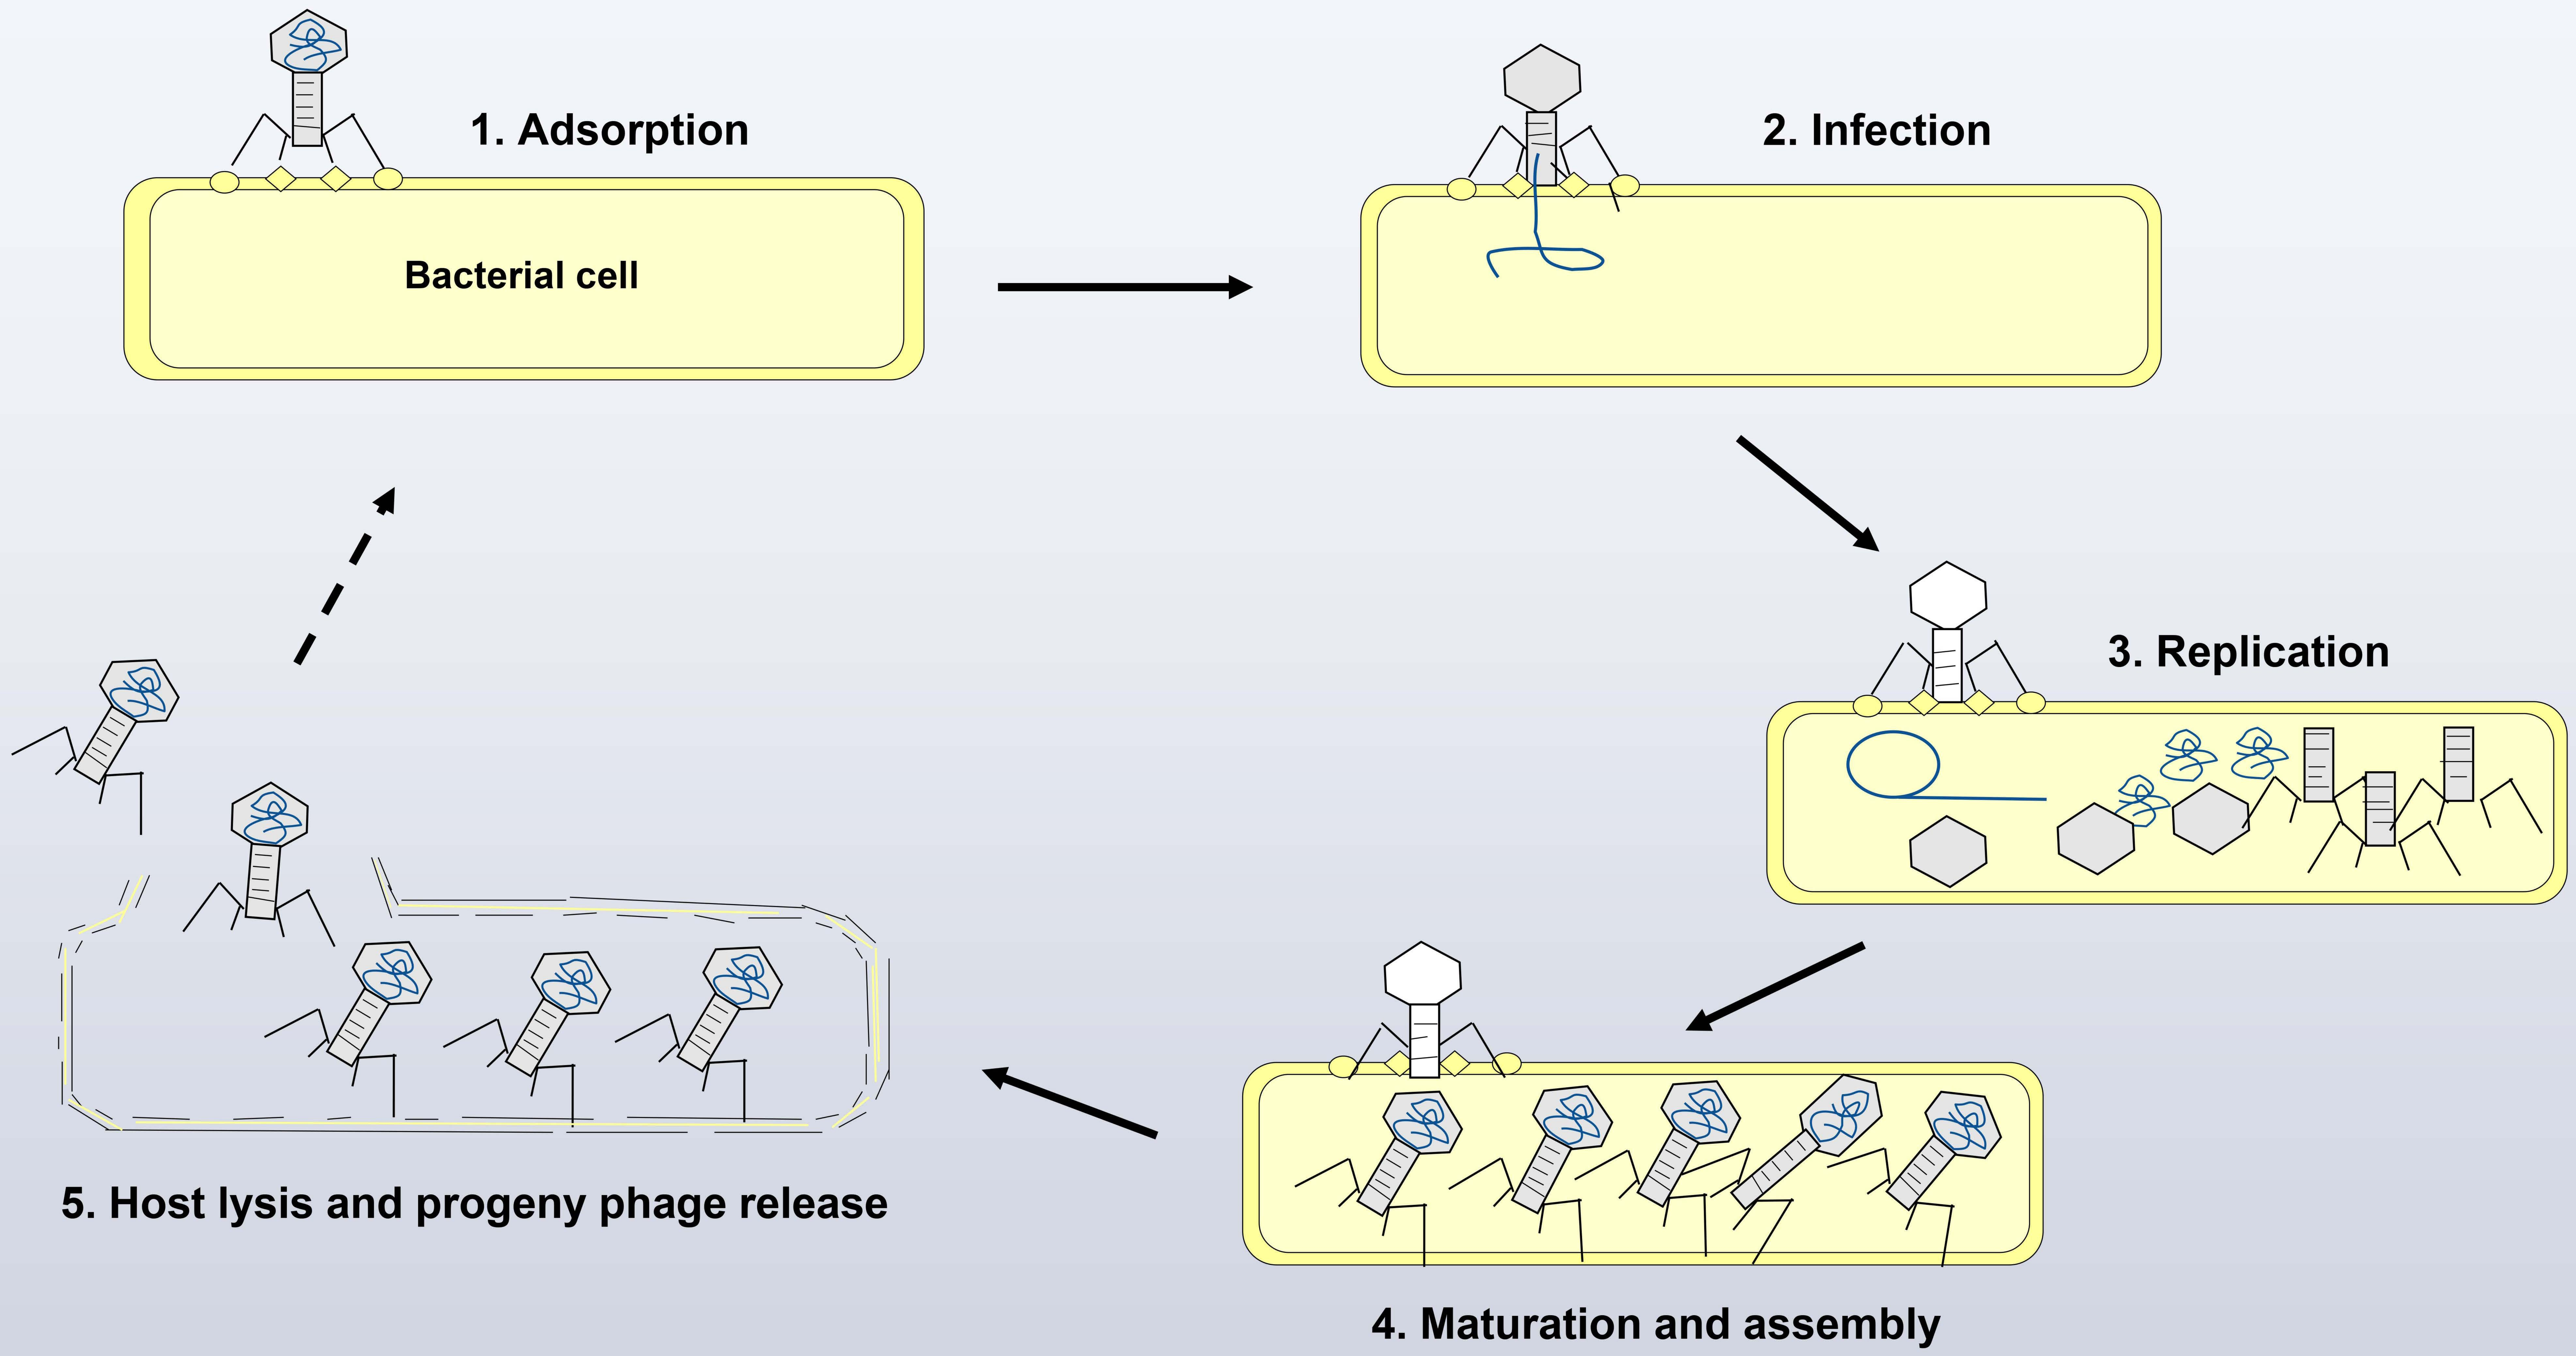

1. Adsorption
Bacterial cell
2. Infection
3. Replication
4. Maturation and assembly
5. Host lysis and progeny phage release

Figure 1. Infection cycle of a bacteriophage. (1) Infection of the bacterial cell starts with adsorption of the bacteriophage to specific receptors on the baterial host. Subsequently, the phage (2) injects its genomic DNA or RNA into the host cell. (3) The phage genome starts replicating and proteins that will stop all regular processes of the bacterial cell are synthesized together with phage components needed to produce progeny phage. (4) Late in infection, phages are assembled inside the bacterial cell after which (5) the bacterial host lysis. To kill *E. coli* on beef, only step 1 and 2 are necessary, as the bacterial cell is already non-viable after these steps.

www.eposterboards.com

Post-Harvest Application of Bacteriophages on Beef as a Natural Intervention Against E. coli O157

Bert de Vegt[1], Silvia Klamert[1], Sander Witte[1], Joël van Mierlo[1]

[1]Micreos Food Safety B.V., Wageningen, The Netherlands

RMC 2018 RECIPROCAL MEAT CONFERENCE

Poster #117

phageguard

Two phage cocktail covers 90% of *E. coli* O157 strains

Strain = Covered **Strain = Not covered**

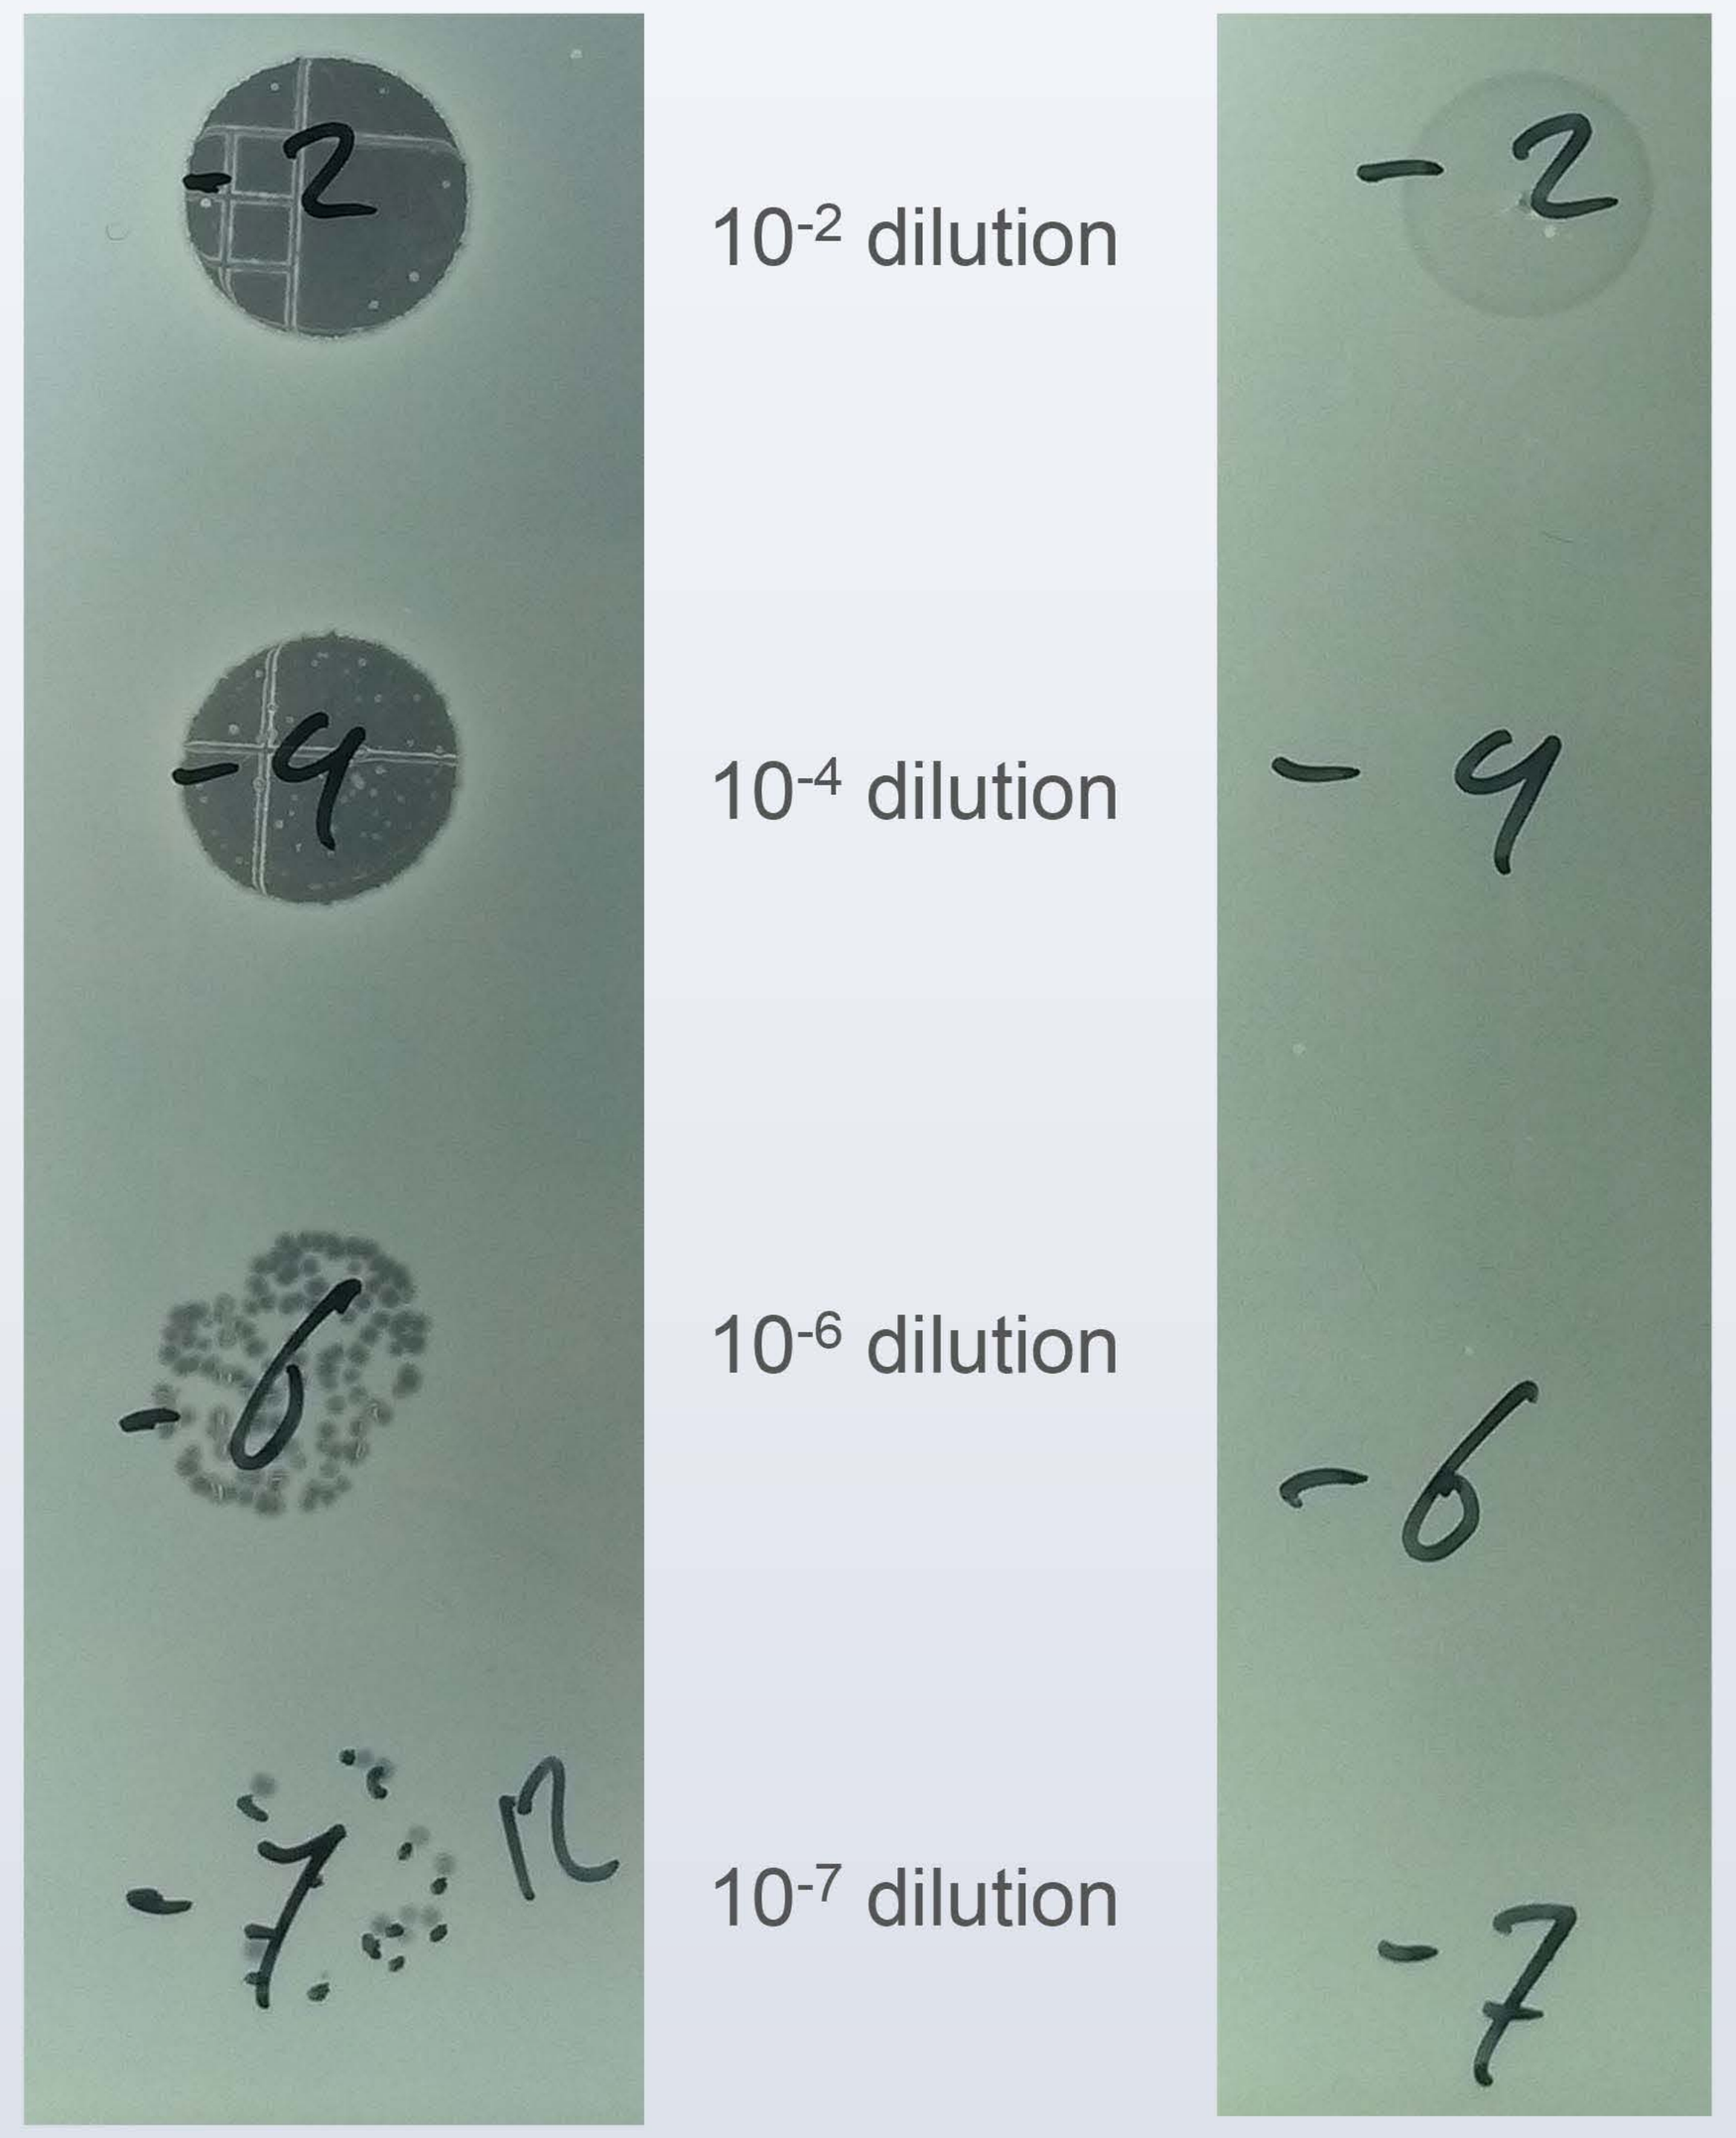
-2
10-2 dilution
-4
10-4 dilution
-6
10-6 dilution
-7
10-7 dilution
-2
-4
-6
-7

Figure 2. The lysis activity of various isolated *E. coli* phages was assessed on 88 *E. coli* O157 strains. Strains were only considered to be covered by a phage if plaques were observed in a high dilution spot (10^{-6} or 10^{-7}).

Table 1. Two phages were selected that showed a complementing hostrange activity, lysing 90 % of all *E. coli* O157 strains tested. A cocktail of these two selected phages was used in all other experiments to evaluate their efficacy on beef contaminated with *E. coli* O157.

Strain type	# strains covered by 2 phage cocktail
E. coli O157 Stx(-)	25/32 (78,1%)
E. coli O157 Stx(+)	54/56 (96,4%)
Total	79/88 (89,7%)

Two phage cocktail reduces *E. coli* O157 on cold beef

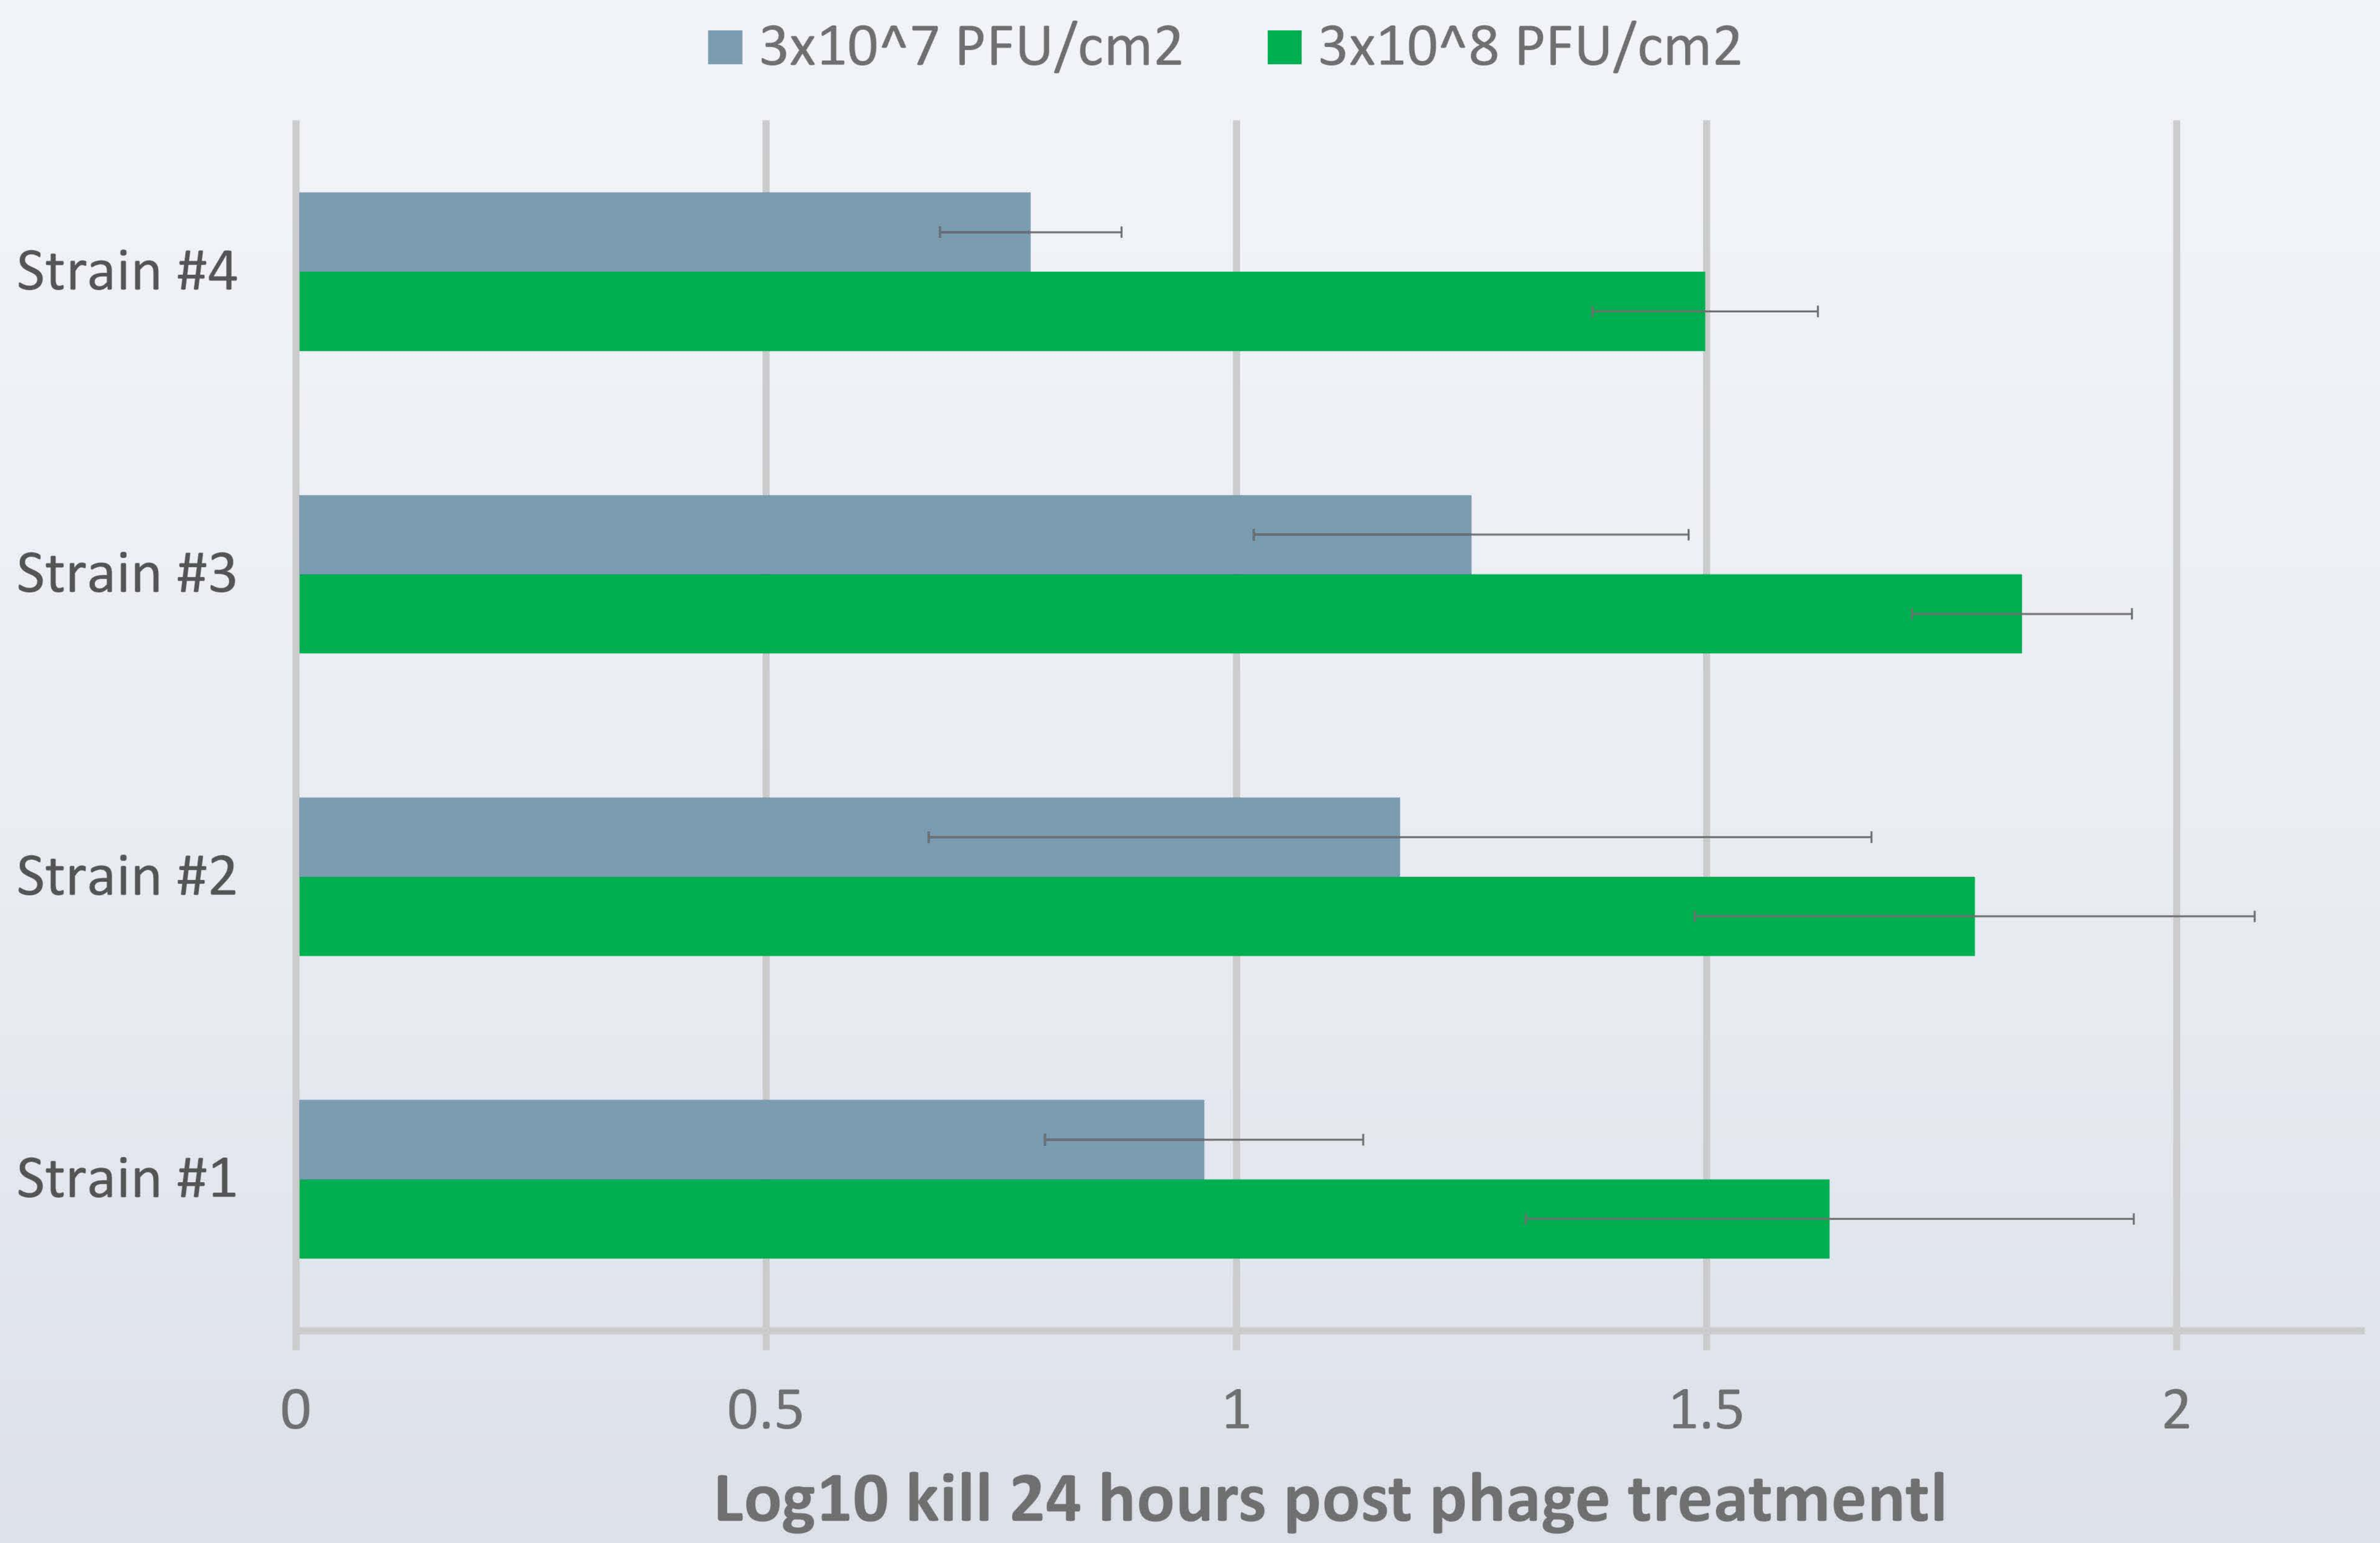
3x10^7 PFU/cm2
3x10^8 PFU/cm2
Strain #4
Strain #3
Strain #2
Strain #1
0
0.5
1
1.5
2
Log10 kill 24 hours post phage treatmentl

Figure 3. For efficacy assays on beef, two cold (4 °C/39.2 °F) beef cuts of 9 cm^2 were contaminated per treatment with *E. coli* O157 at a rate of 1 x 10^5 cfu/cm^2. Subsequently, samples were treated with 3 x 10^7 or 3 x 10^8 Plaque Forming Units (PFU)/cm^2 by applying 5 µL/cm^2 of a cocktail containing two selected phages. Controls were treated with similar volumes of tap water. Beef samples were then incubated for 24 hours at 4 °C, after which bacteria were retrieved for enumeration. Bacterial reductions on phage treated samples were calculated relative to tap water treated controls. Reductions of four different *E. coli* O157 strains obtained in three independent experiments were used for statistical analysis (Unpaired t-test, all reductions displayed have P<0.05).

Figure 4 and Figure 5. Two separate time trials were done in which beef samples were contaminated and treated as described above. In one time trial, beef samples were stored at 4 °C during the complete experiment (**Figure 4**). In the second time trial, all samples were initially stored at 4 °C for 24 hours, after which all remaining samples were transferred to an abusive temperature of 20 °C (68.0 °F) for the remainder of the experiment (**Figure 5**). For both time trials, two beef samples per treatment were retrieved for bacterial enumeration at 2, 6, 24, 30, 48, and 54 hours post phage treatment. Results were confirmed in three independent experiments on which statistical analysis per time point was done (Unpaired t-test, all time points have P<0.05).

Maximum kill of *E. coli* O157 after 6 hours dwell time

Figure 4

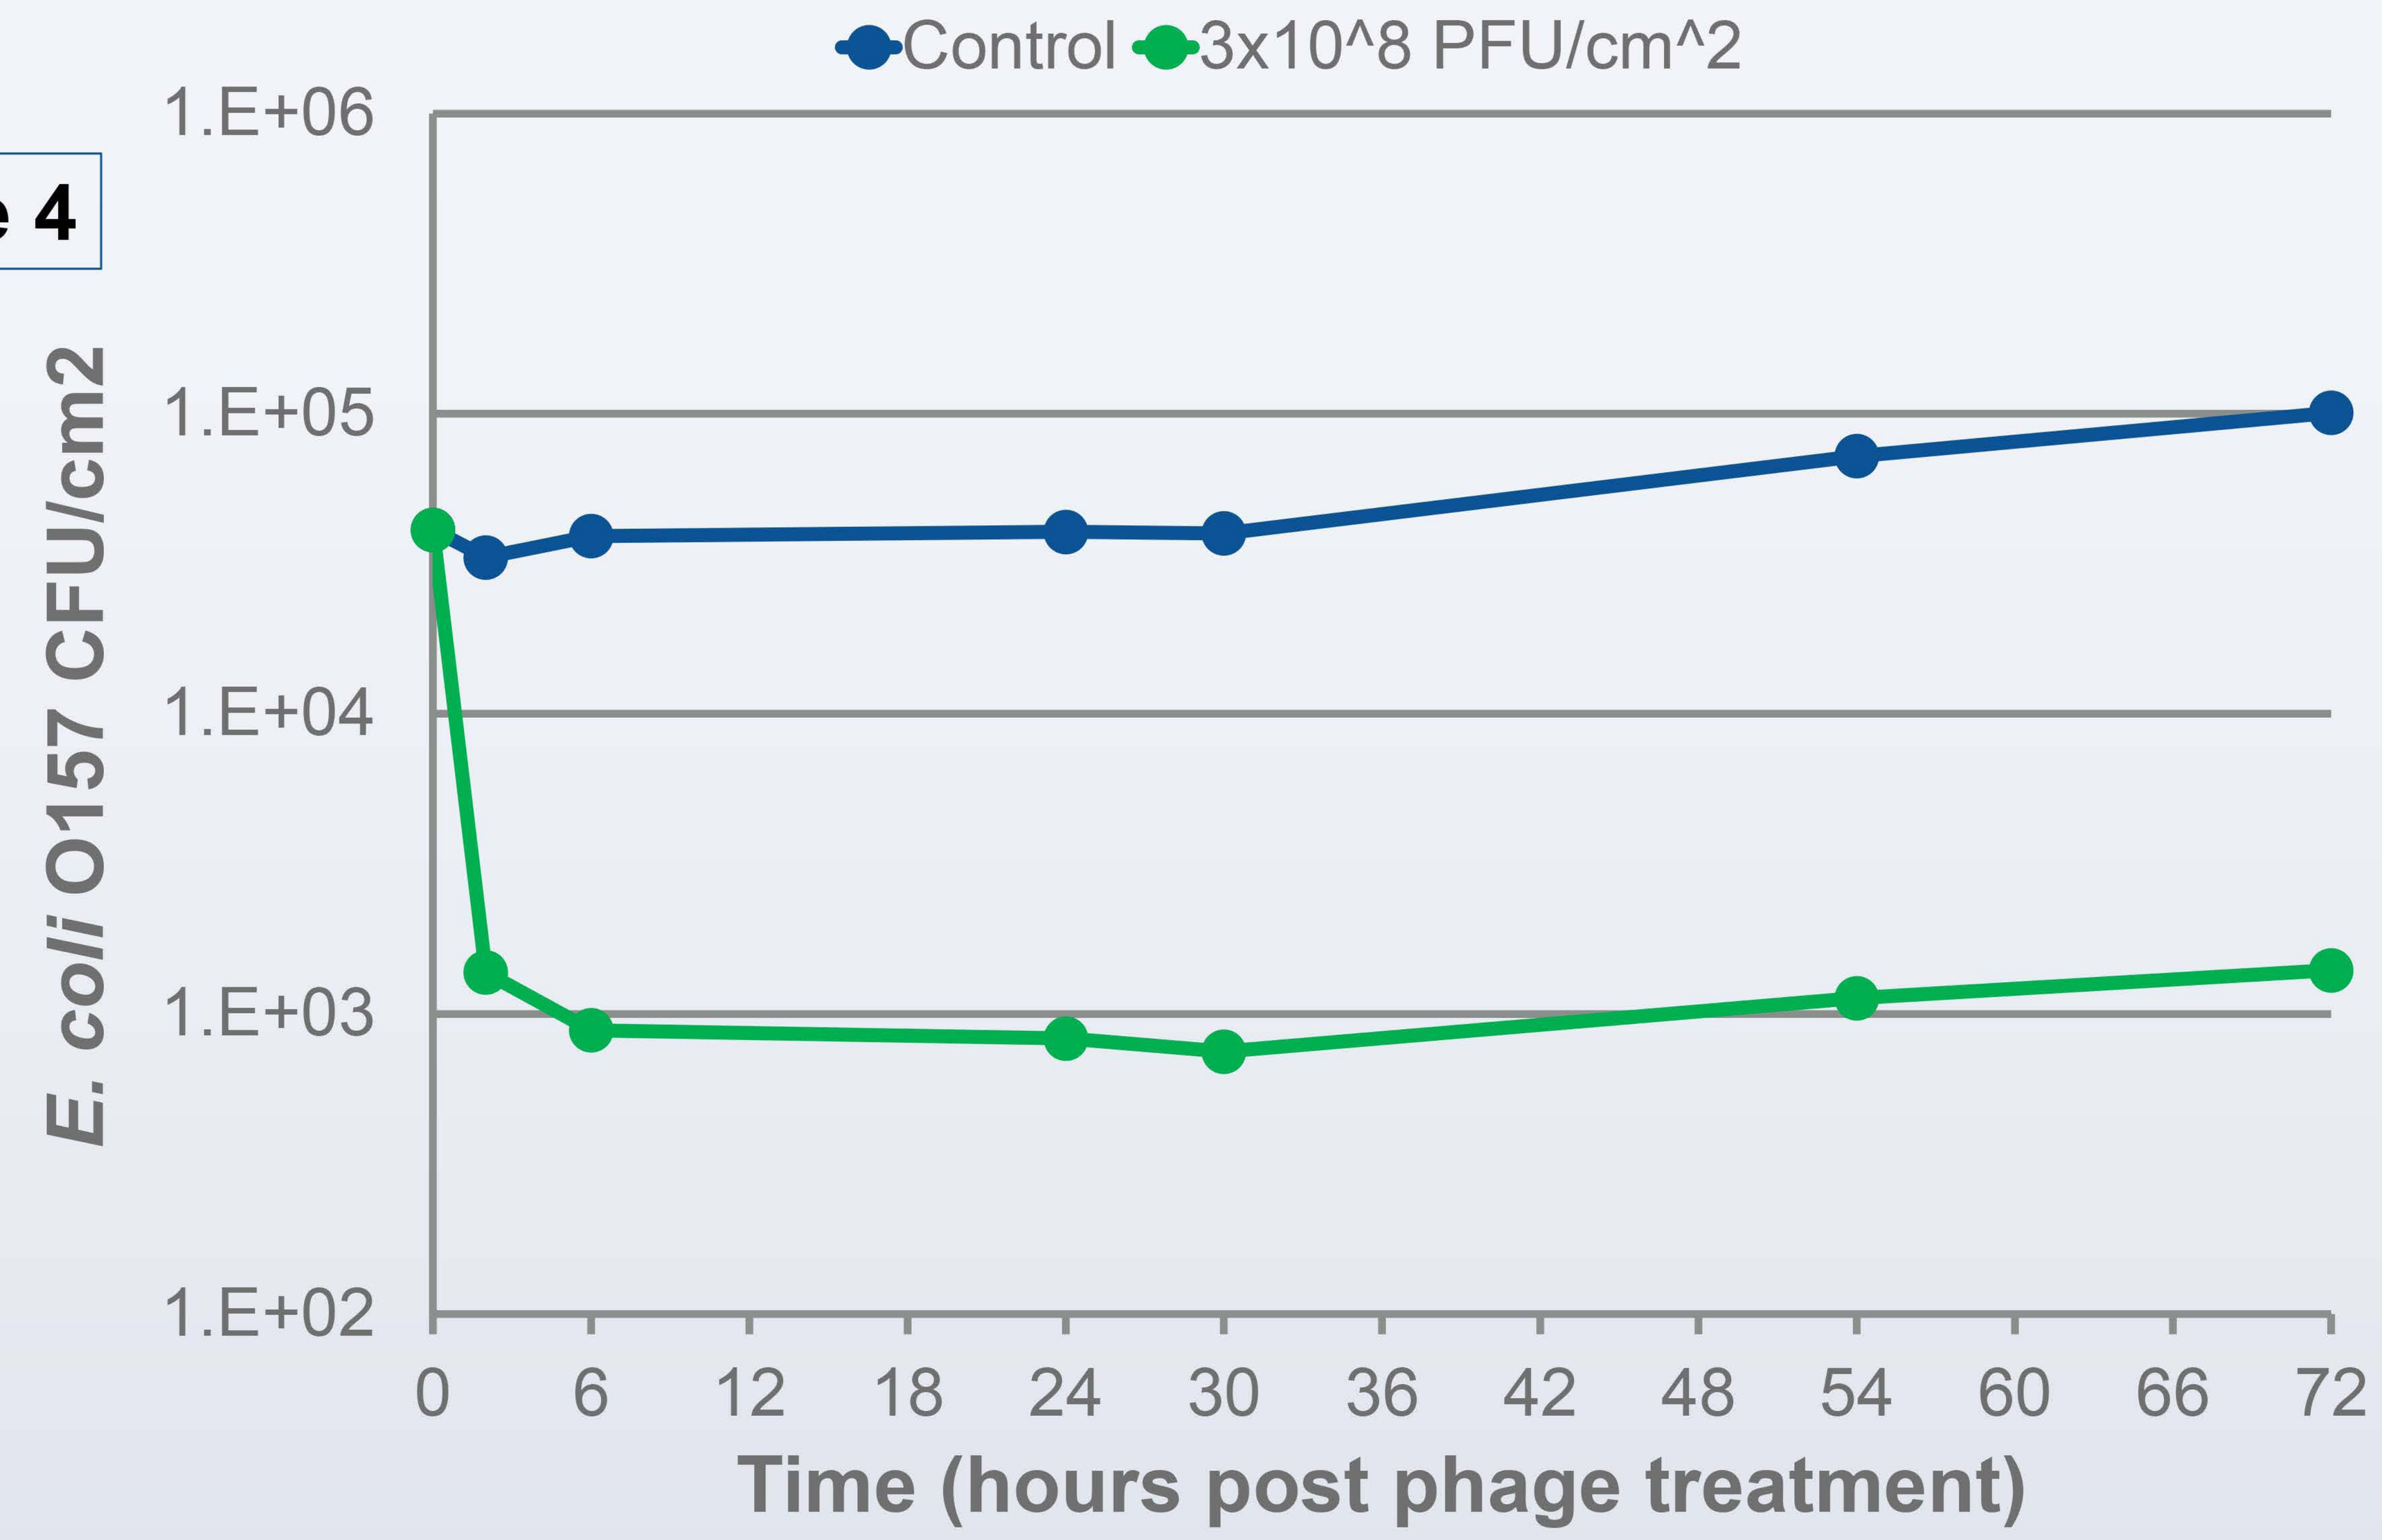
Control
3x10^8 PFU/cm^2
1.E+06
1.E+05
1.E+04
1.E+03
1.E+02
E. coli O157 CFU/cm2
0
6
12
18
24
30
36
42
48
54
60
66
72
Time (hours post phage treatment)

Two phage cocktail against *E. coli* O157 is processing aid

Figure 5

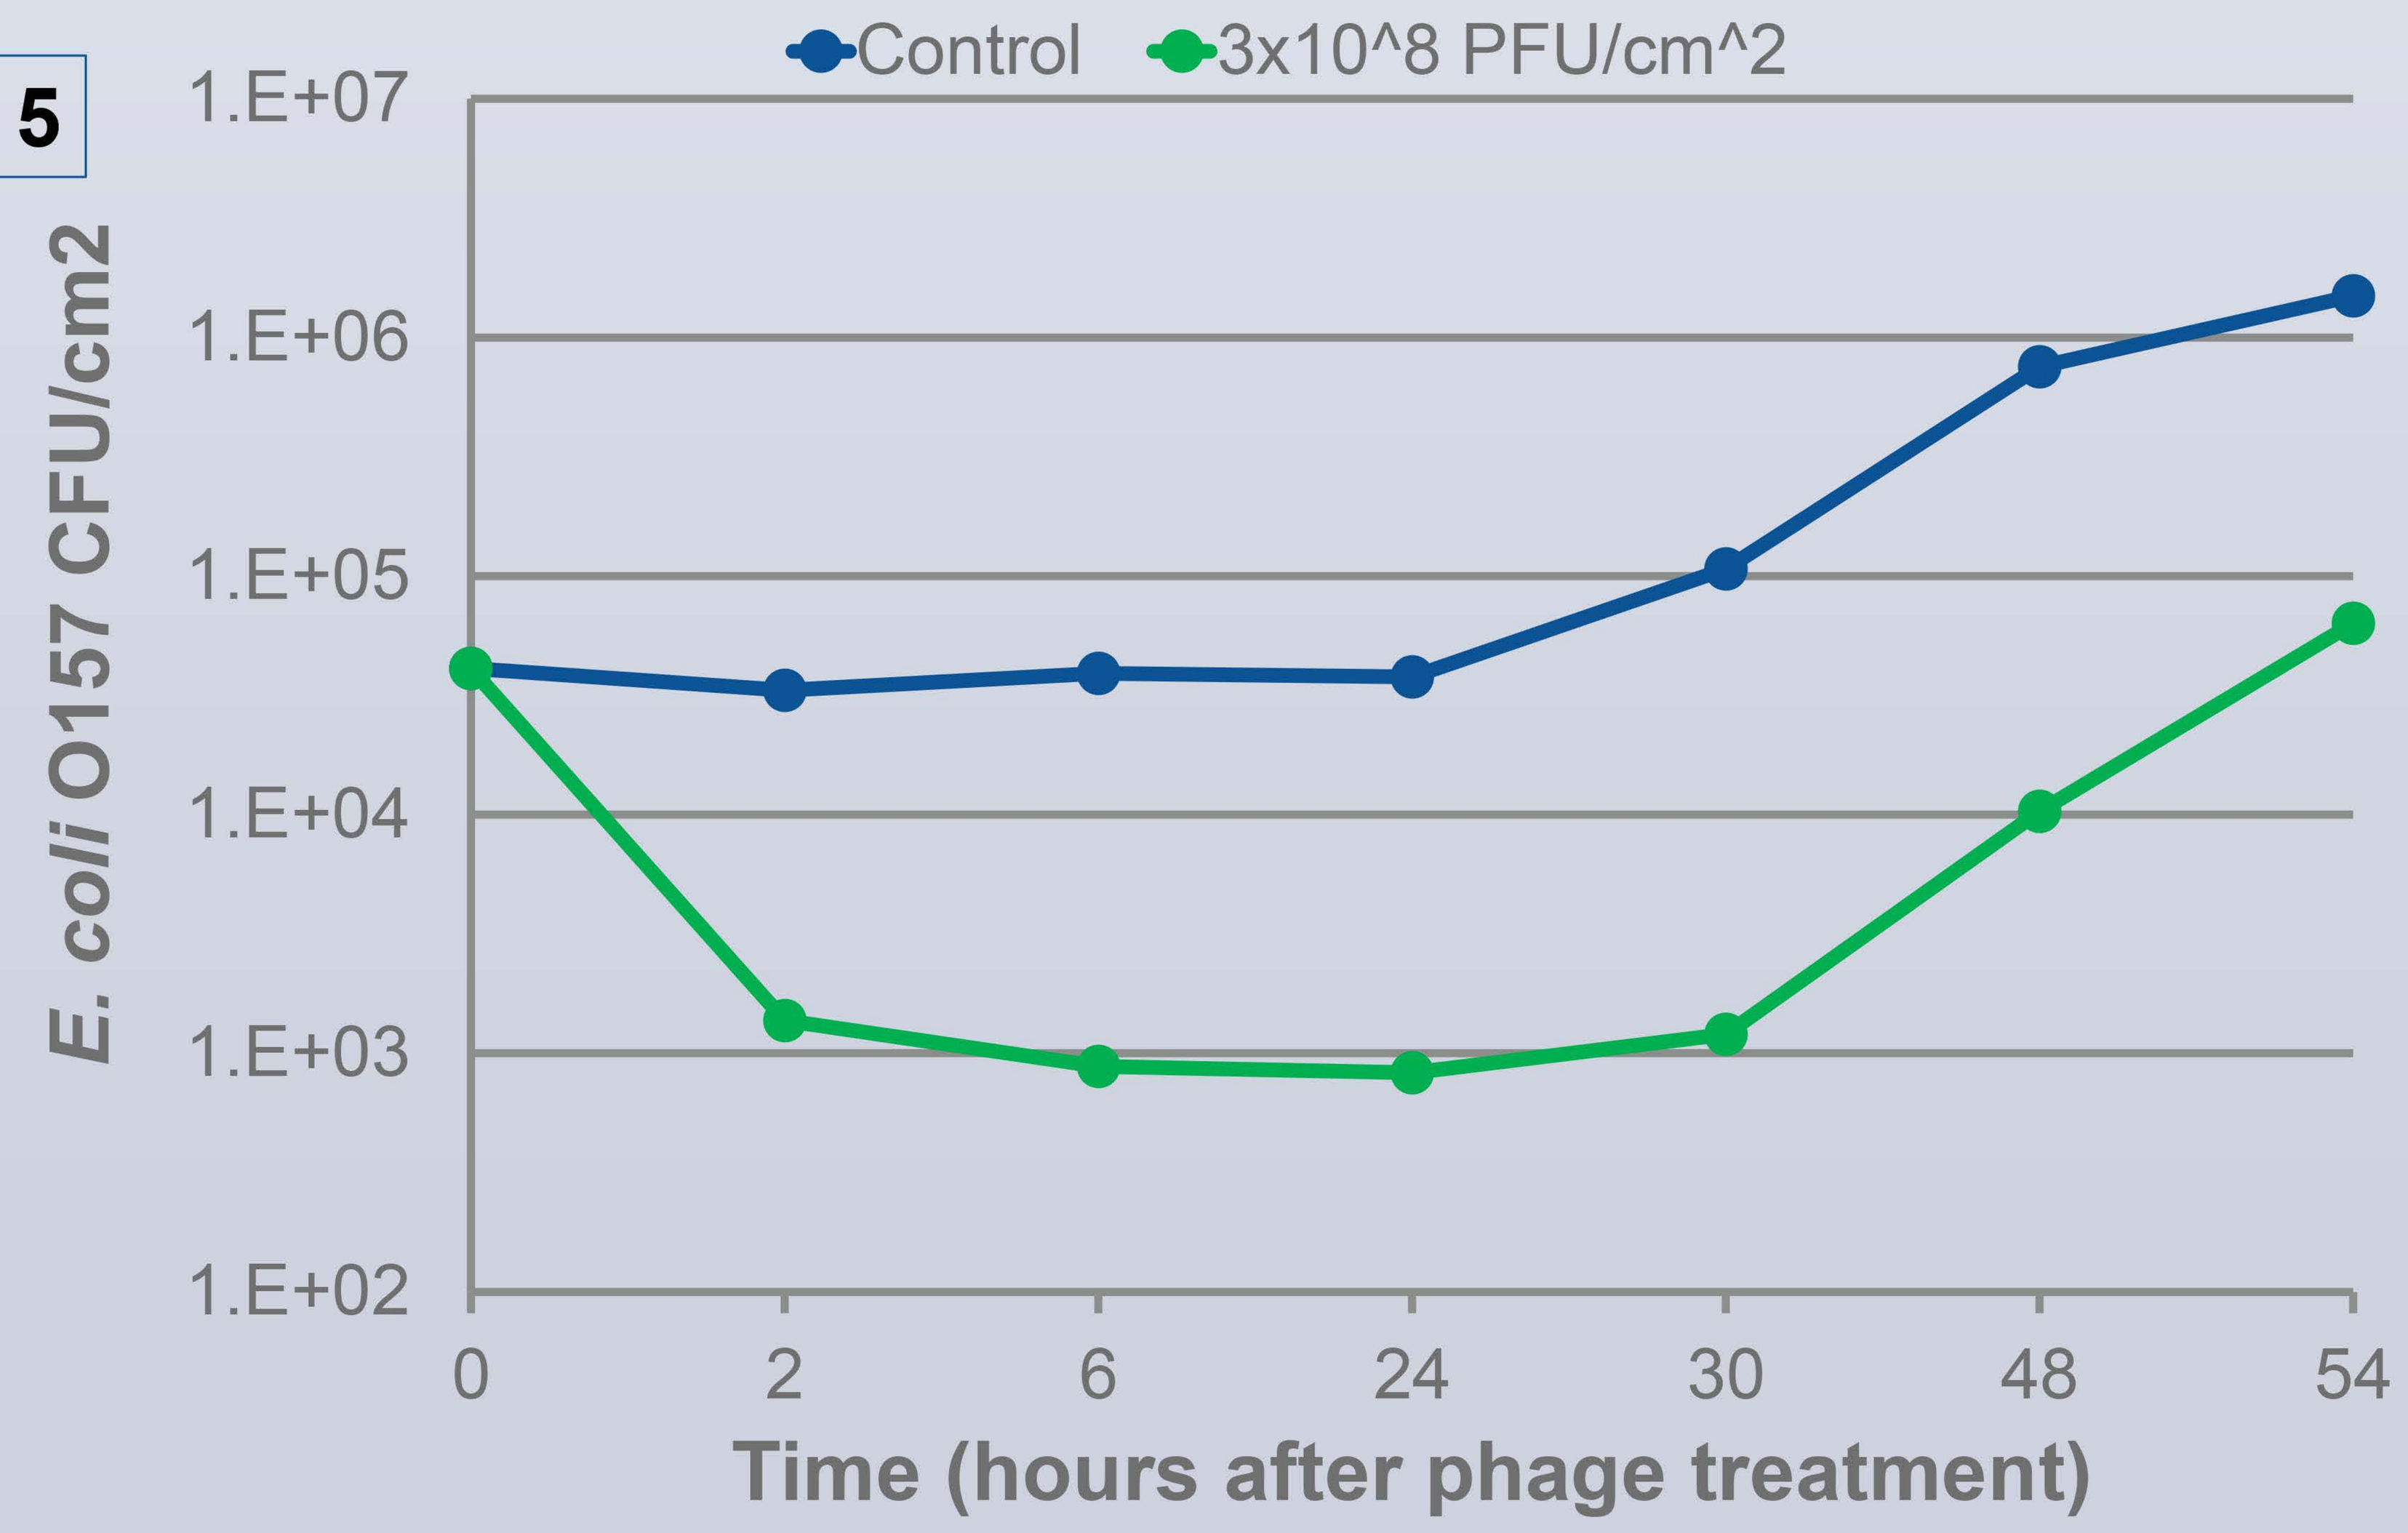
Control
3x10^8 PFU/cm^2
1.E+07
1.E+06
1.E+05
1.E+04
1.E+03
1.E+02
E. coli O157 CFU/cm2
0
2
6
24
30
48
54
Time (hours after phage treatment)

www.eposterboards.com